#2084

BI 685509 improves hepatic function in subjects with Child–Pugh A cirrhosis and a liver stiffness measurement of >15 kPa: Results from the HepQuant SHUNT test

Eric Lawitz[1], Judith Ertle[2], Corinna Schoelch[3], Isabella Gashaw[4], Steve M. Helmke[5], Gregory T. Everson[5]

[1]Texas Liver Institute, University of Texas, San Antonio, TX, USA; [2]Boehringer Ingelheim International GmbH, Ingelheim am Rhein, Germany; [3]Boehringer Ingelheim Pharma GmbH & Co. KG, Biberach, Germany; [4]Boehringer Ingelheim Pharma GmbH & Co. KG, Ingelheim am Rhein, Germany; [5]HepQuant LLC, Greenwood Village, CO, USA

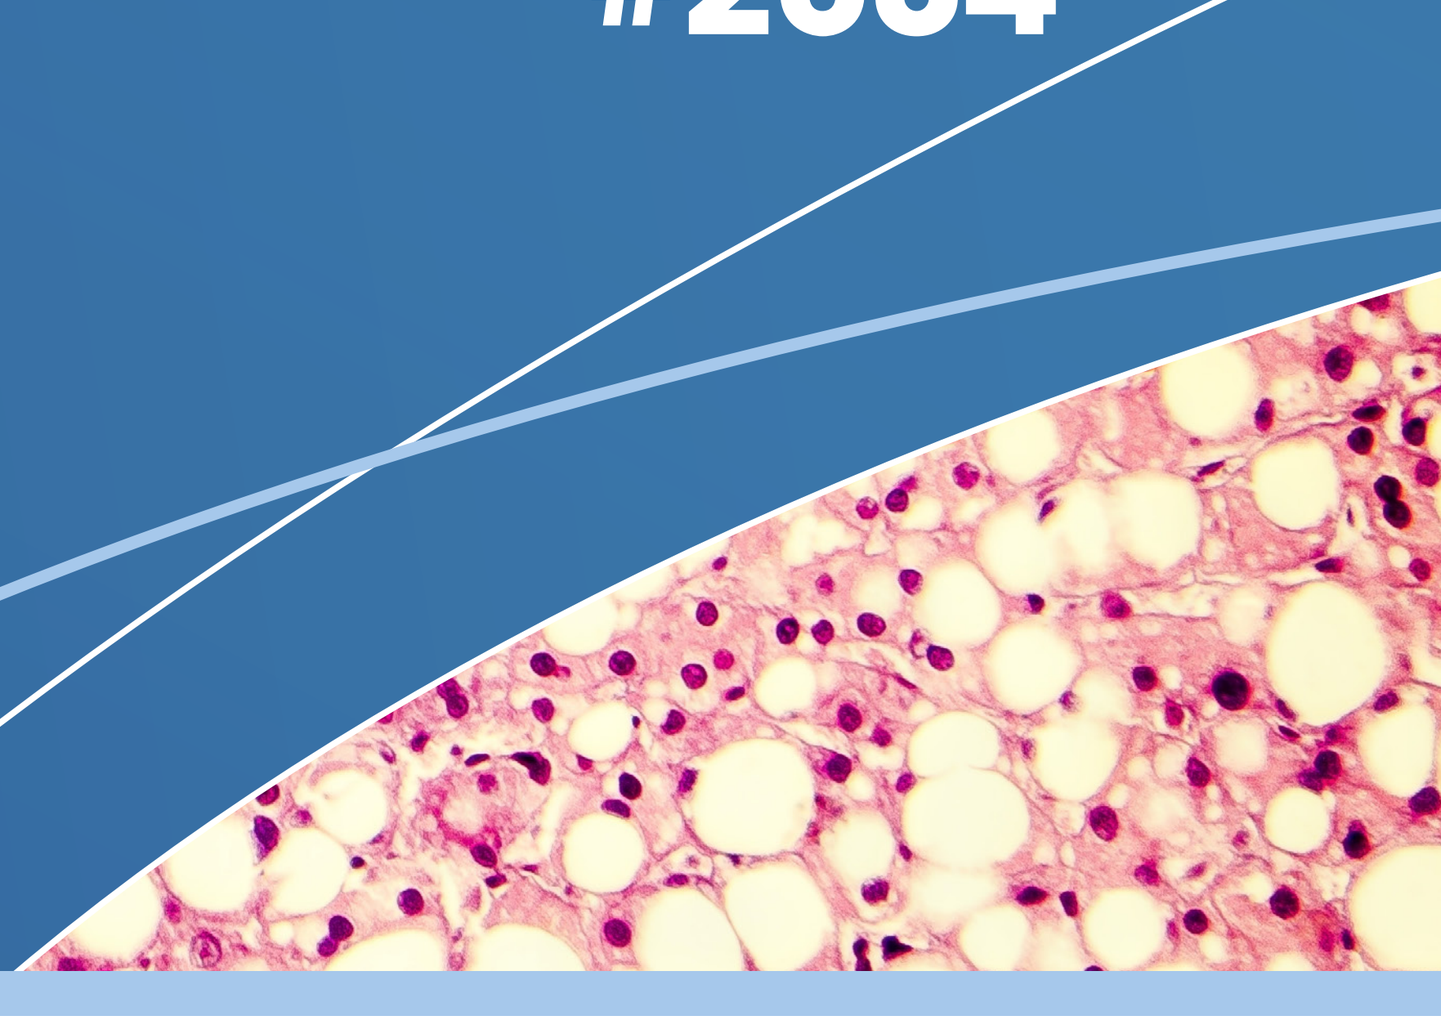

Background and aims:

- BI 685509 is a nitric oxide (NO)-independent activator of soluble guanylate cyclase (sGC), which catalyzes production of cyclic guanosine monophosphate (cGMP)[1,2]
- cGMP stimulates hepatic vasodilation,[3] thereby increasing sinusoidal perfusion;[4] dysfunction of the NO-sGC-cGMP pathway in cirrhosis increases portal pressure and leads to portal hypertension[5–7]
- The HepQuant SHUNT test measures effective sinusoidal perfusion as a result of the flow-dependent hepatic clearance of cholate (CA) from the systemic and portal circulations[8]
- The test quantifies portal-systemic shunting (SHUNT) and first-pass hepatic extraction (1 – SHUNT) and can identify the risk of well-compensated patients to develop complications of cirrhosis, including portal hypertension, in a minimally invasive manner[8,9]
- In this subgroup analysis of a Phase Ib study (NCT03842761), the HepQuant SHUNT test was used to measure the hepatic effects of BI 685509 in subjects with Child–Pugh A (CP-A) cirrhosis

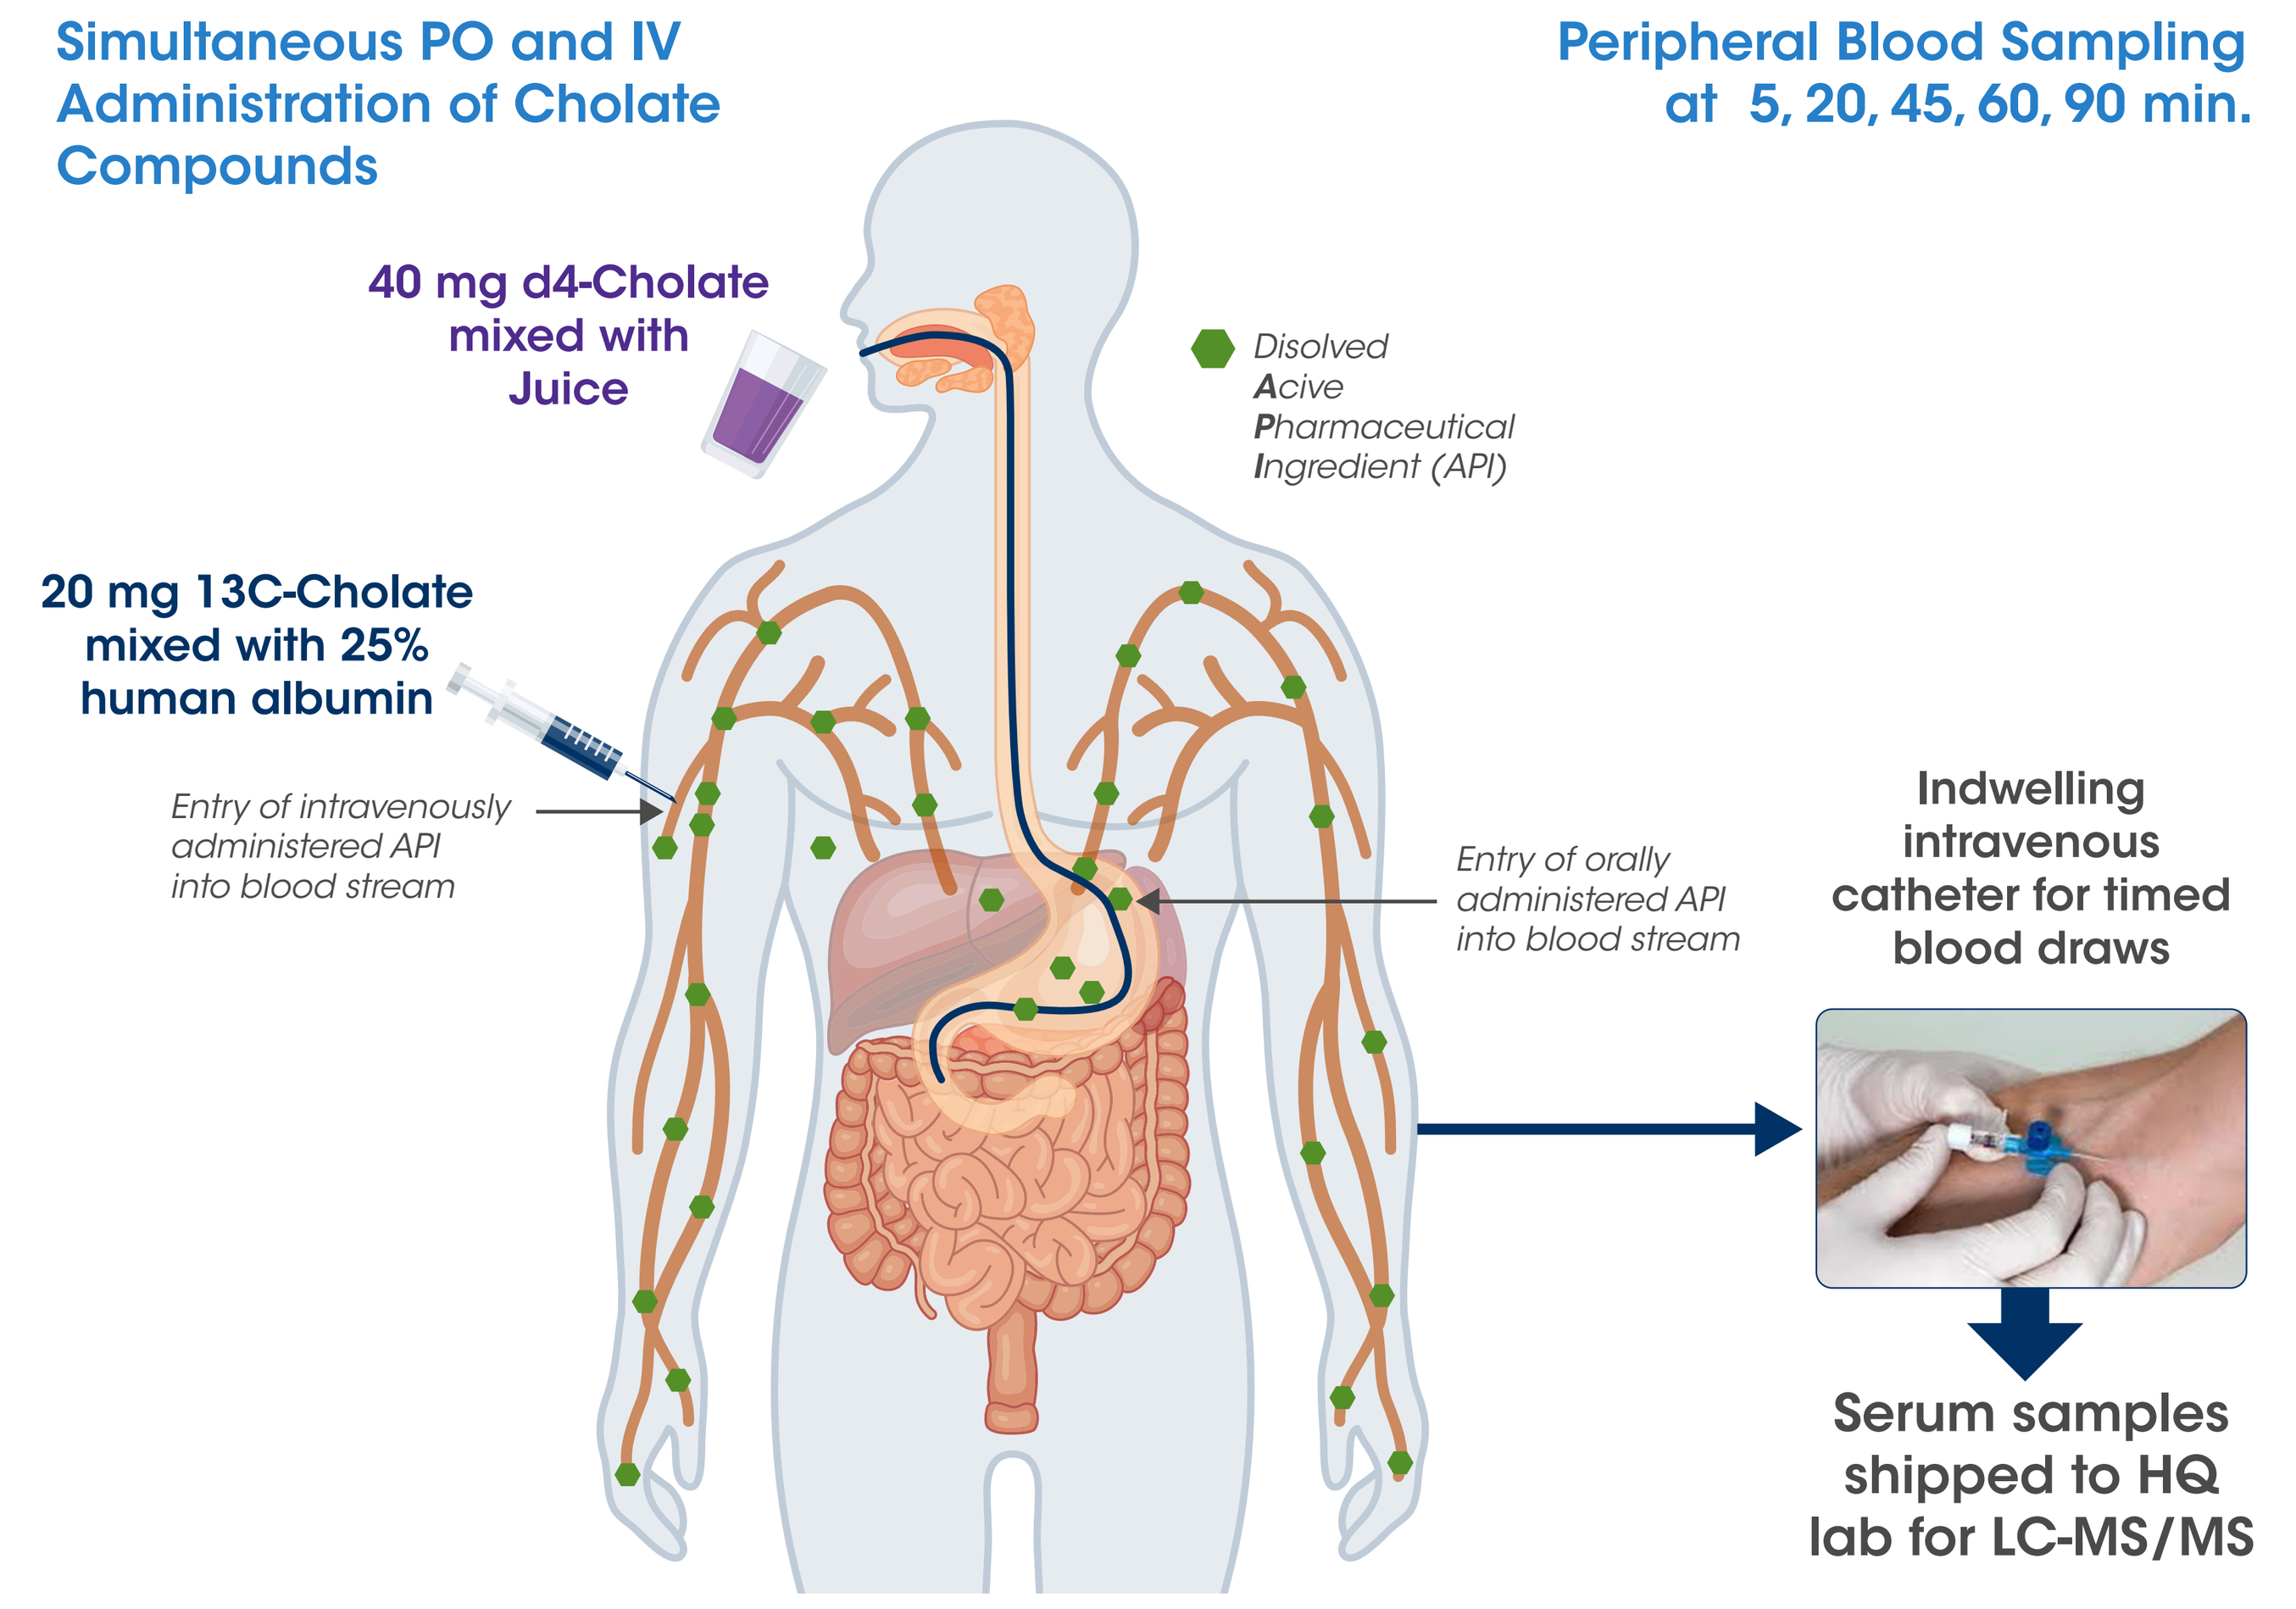
Simultaneous PO and IV Administration of Cholate Compounds
Peripheral Blood Sampling at 5, 20, 45, 60, 90 min.
40 mg d4-Cholate mixed with Juice
Disolved Acive Pharmaceutical Ingredient (API)
20 mg 13C-Cholate mixed with 25% human albumin
Entry of intravenously administered API into blood stream
Entry of orally administered API into blood stream
Indwelling intravenous catheter for timed blood draws
Serum samples shipped to HQ lab for LC-MS/MS

API, active pharmaceutical ingredient; HQ, HepQuant; IV, intravenous; LC-MS/MS, liquid chromatography tandem mass spectrometry; PO, per os.

Methods:

- This randomized, placebo-controlled, double-blind (within dose groups), parallel-group, dose-escalation study investigated three dosing schemes of BI 685509 (**Figure 1**) in subjects with CP-A cirrhosis, liver stiffness >15 kPa, and mean arterial pressure ≥85 mmHg for 28 days
 - The maximum tested dosages after titration were 1 mg twice daily (BID), 2 mg BID, and 3 mg BID
 - Further information on this study is being presented at AASLD 2021 (poster presentation #2092)
- HepQuant SHUNT tests were performed at baseline and on Days 11 and 27 of treatment, including an intravenous injection of 13C CA, oral administration of d4 CA, and five blood samples drawn over 90 minutes
- Serum was analyzed and hepatic filtration rates (HFRs), SHUNT (systemic/portal HFR), first-pass extraction (1 – SHUNT), STAT (d4-CA at 60 minutes), disease severity index (DSI), and hepatic reserve (HR) were calculated

Figure 1. Study dose groups

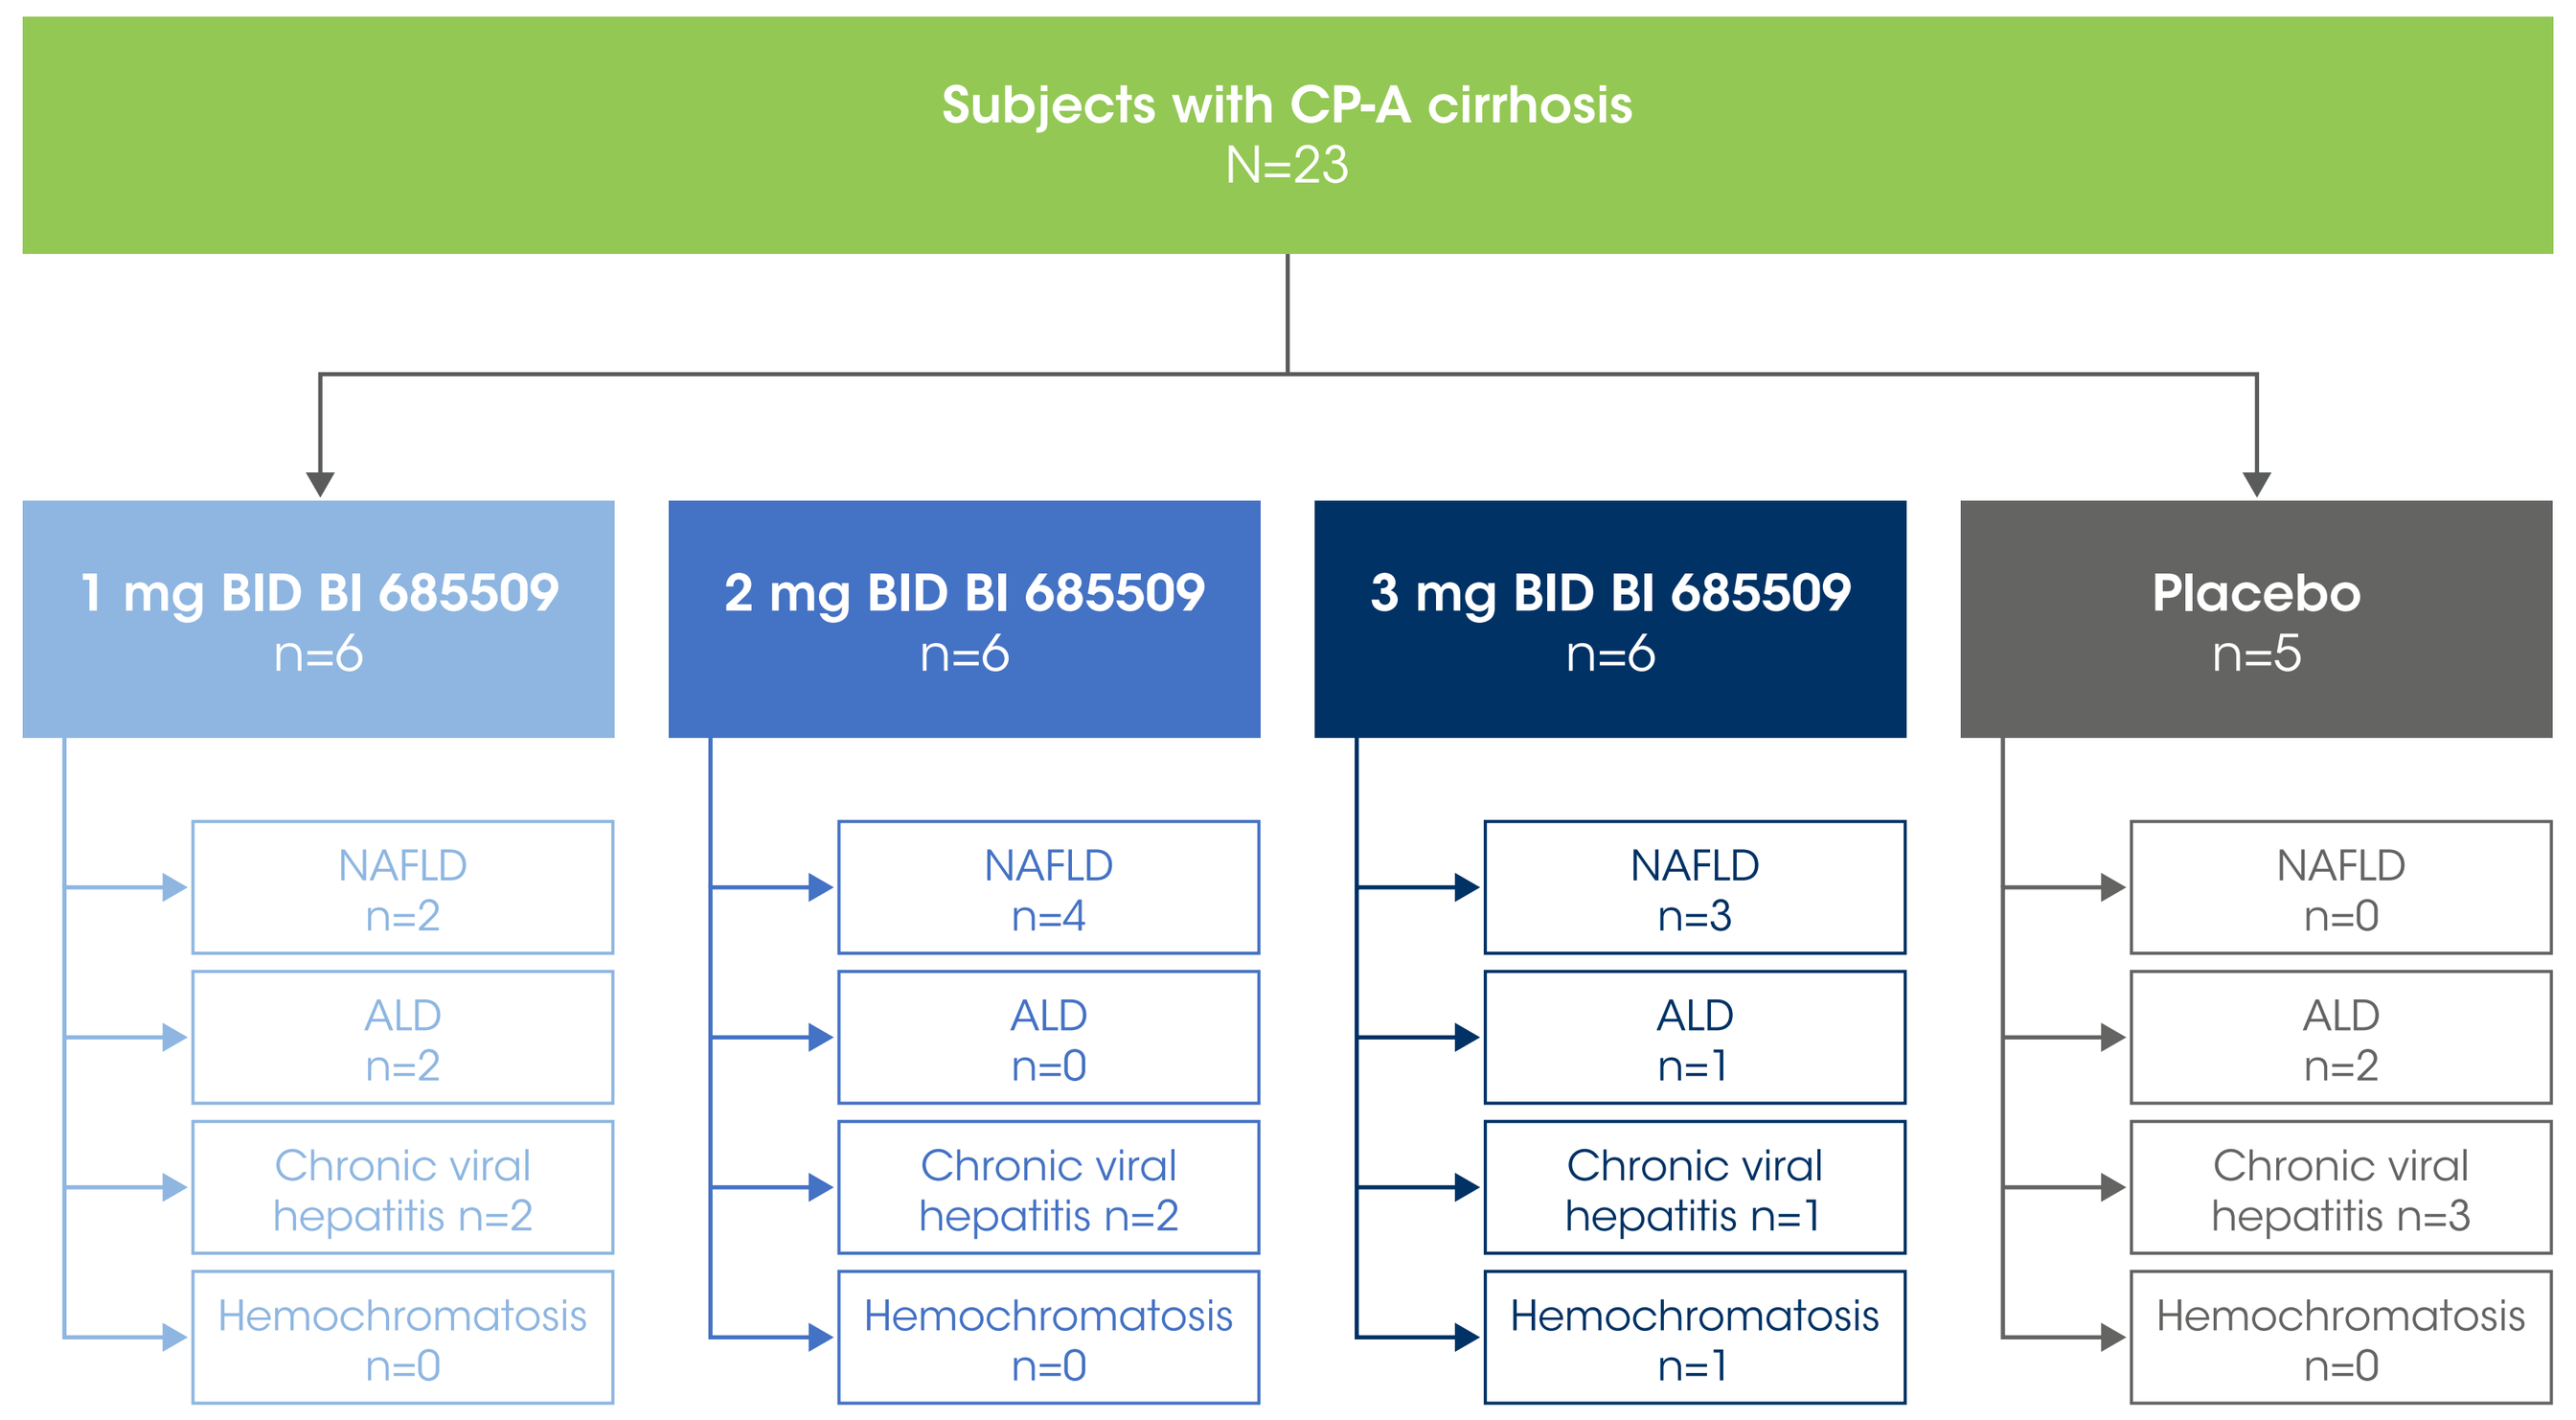
Subjects with CP-A cirrhosis
N=23
1 mg BID BI 685509
n=6
2 mg BID BI 685509
n=6
3 mg BID BI 685509
n=6
Placebo
n=5
NAFLD n=2
ALD n=2
Chronic viral hepatitis n=2
Hemochromatosis n=0
NAFLD n=4
ALD n=0
Chronic viral hepatitis n=2
Hemochromatosis n=0
NAFLD n=3
ALD n=1
Chronic viral hepatitis n=1
Hemochromatosis n=1
NAFLD n=0
ALD n=2
Chronic viral hepatitis n=3
Hemochromatosis n=0

ALD, alcoholic liver disease; BID, twice daily; CP-A, Child–Pugh A; NAFLD, non-alcoholic fatty liver disease.

Results:

Subject demographics were similar between dose groups

- All patients included in this trial were white
- Two subjects progressed from CP-A to CP-B by Day 28 (n=1 each in the 1 mg BID and 3 mg BID groups)

Table 1. Patient demographics and disease characteristics

Characteristic	1 mg BID (n=6)	2 mg BID (n=6)	3 mg BID (n=6)	Placebo (n=5)	Total (n=23)
Sex, male, n (%)	3 (50.0)	2 (33.3)	4 (66.7)	2 (40.0)	11 (47.8)
Age, years, mean (SD)	59.5 (6.0)	62.2 (5.2)	61.8 (7.3)	58.2 (7.6)	60.5 (6.3)
Bodyweight, kg, mean (SD)	82.0 (13.0)	85.1 (12.4)	93.5 (15.1)	76.6 (11.6)	84.6 (13.7)
BMI, kg/m², mean (SD)	30.9 (5.1)	32.9 (5.7)	33.3 (5.7)	28.1 (3.4)	31.4 (5.2)
MELD, mean (SD)	8.2 (2.1)	7.3 (1.4)	7.7 (1.2)	7.8 (1.1)	7.7 (1.5)
Liver stiffness, kPa, mean (SD)	29.3 (10.7)	23.0 (10.9)	25.1 (2.1)	27.2 (8.3)	26.1 (8.5)
Spleen stiffness, kPa, mean (SD)	71.6 (19.4)	51.7 (19.5)	63.4 (29.9)	55.8 (27.5)	60.4 (23.1)
MAP, mmHg, (SD)	95.5 (10.9)	100.8 (14.7)	99.2 (8.2)	94.9 (15.0)	97.7 (11.8)
Cirrhosis etiology, n (%)					
NAFLD	2 (33.3)	4 (66.7)	3 (50.0)	0	9 (39.1)
ALD	2 (33.3)	0	1 (16.7)	2 (40.0)	5 (21.7)
Chronic viral hepatitis	2 (33.3)	2 (33.3)	1 (16.7)	3 (60.0)	8 (34.8)
Hemochromatosis	0	0	1 (16.7)	0	1 (4.3)

ALD, alcoholic liver disease; BID, twice daily; BMI, body mass index; MAP, mean arterial pressure; MELD, model for end-stage liver disease; NAFLD, non-alcoholic fatty liver disease; SD, standard deviation.

First-pass extraction of d4-CA increased in a dose-dependent manner with BI 685509

Figure 2. Change in the first pass extraction % of d4-CA (1 – SHUNT %) from the portal inflow

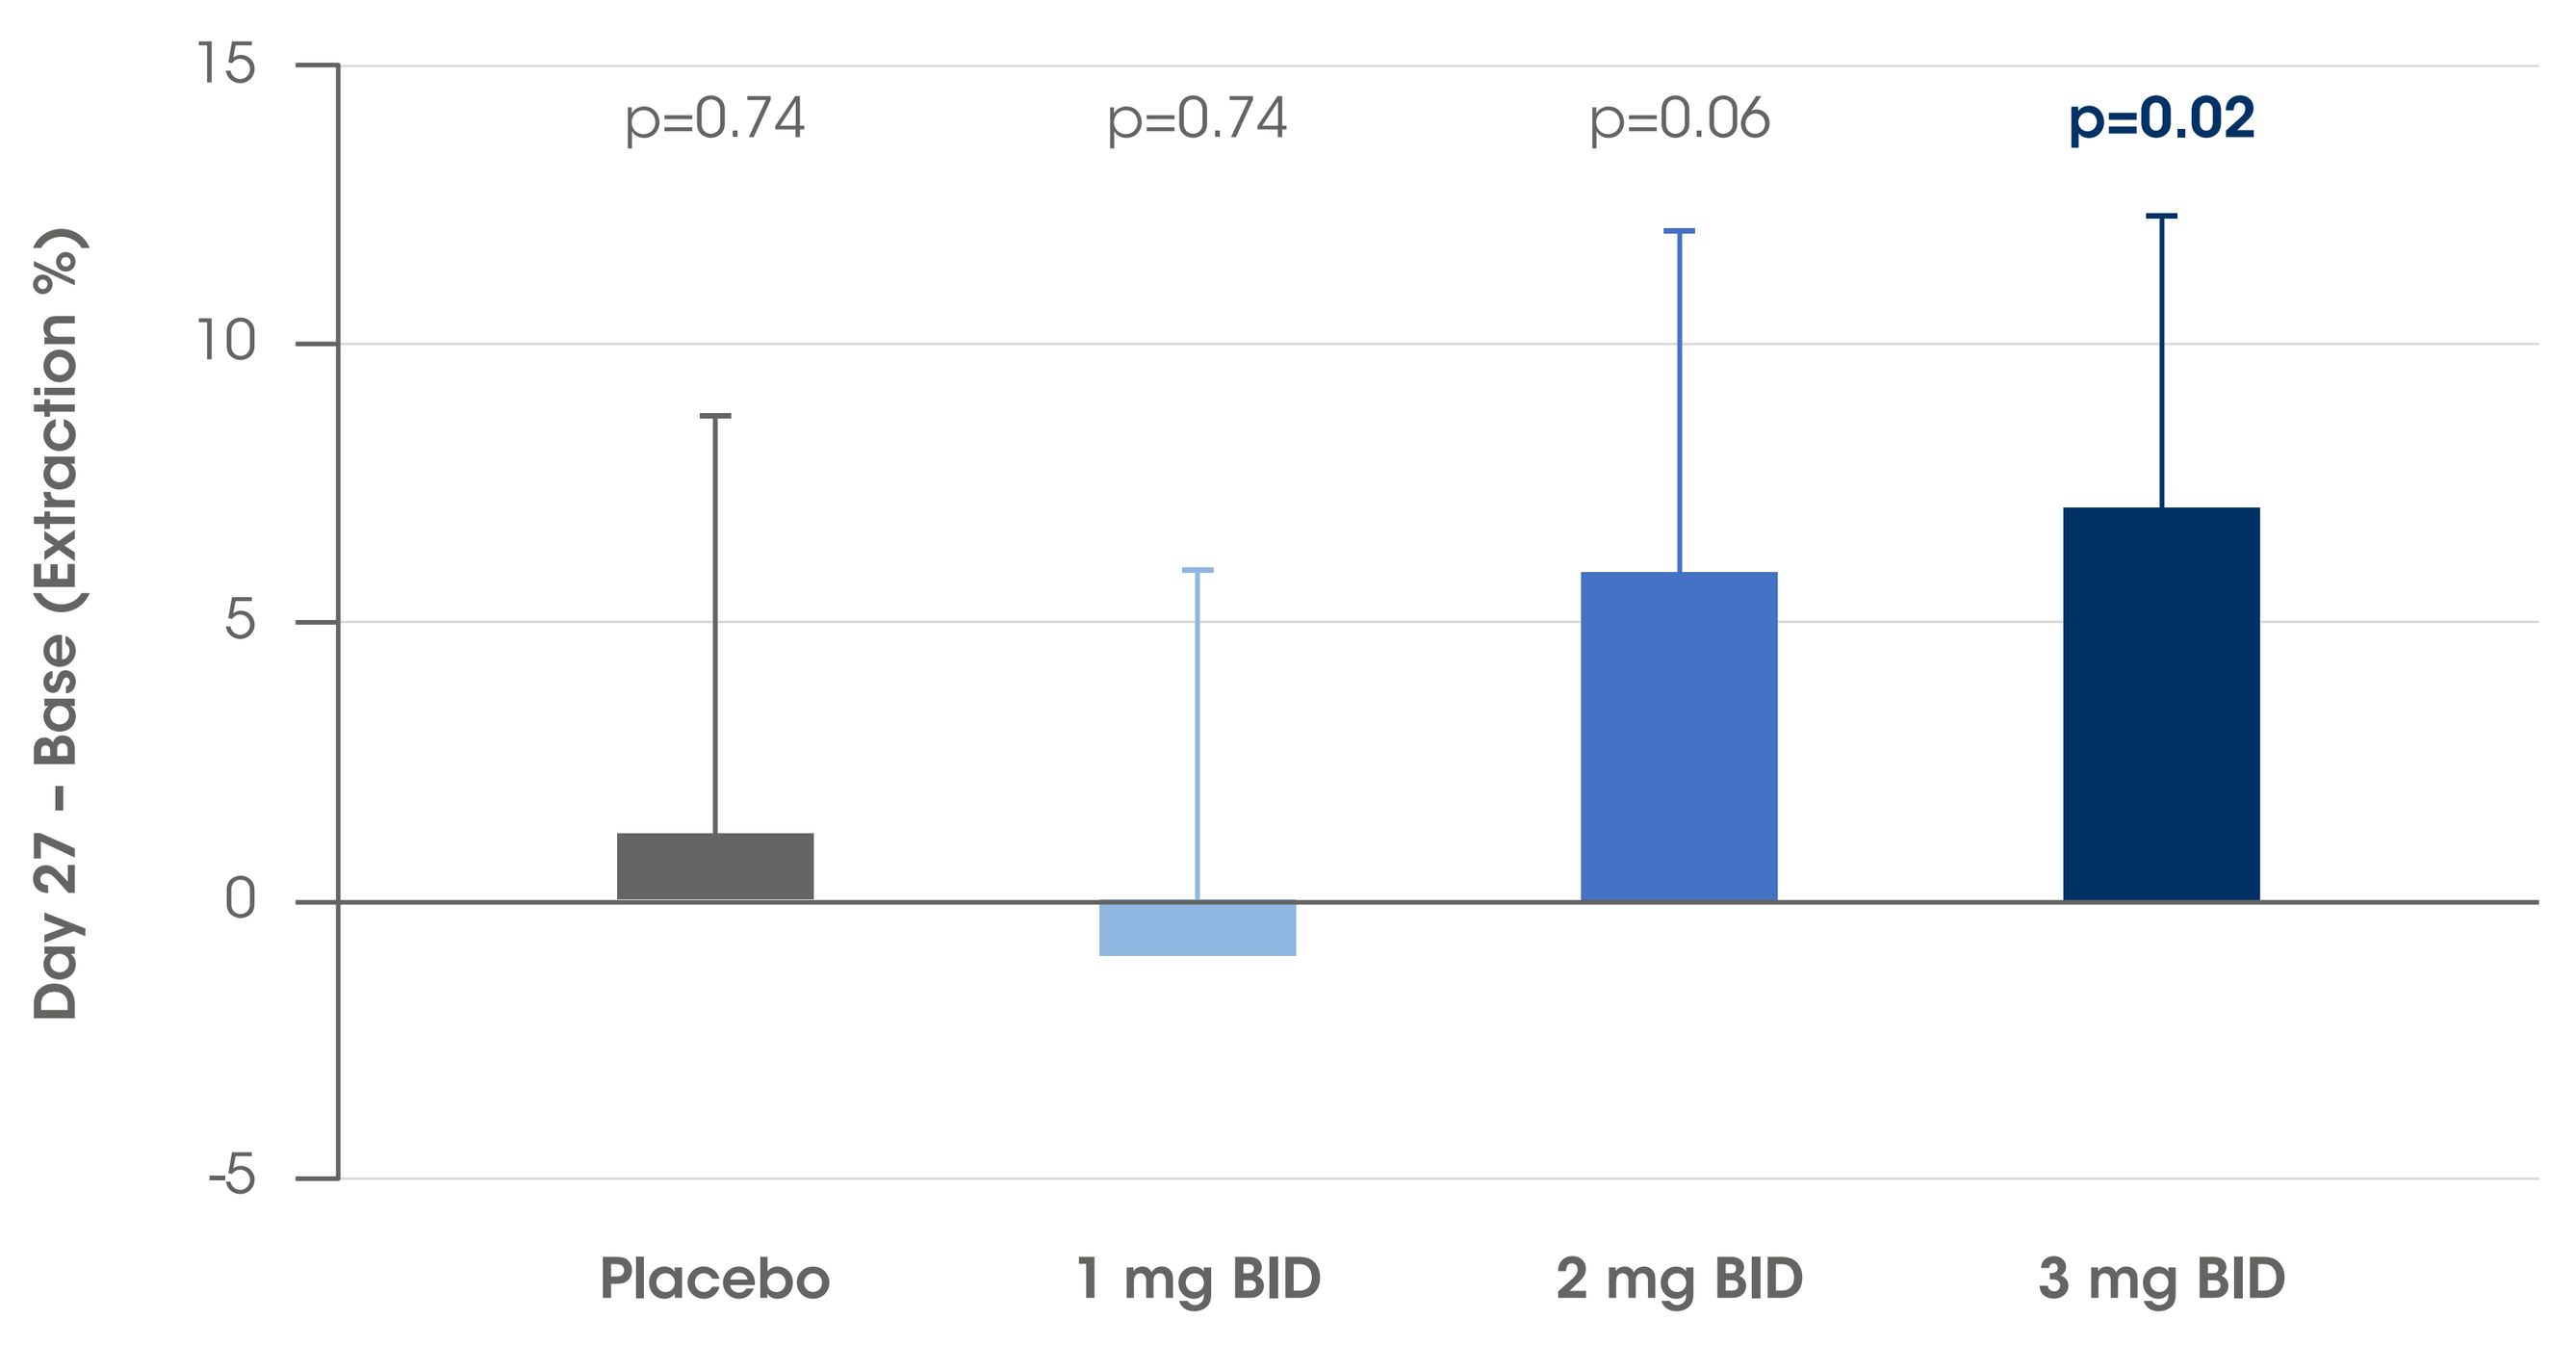
15
10
5
0
-5
Day 27 – Base (Extraction %)
p=0.74
p=0.74
p=0.06
p=0.02
Placebo
1 mg BID
2 mg BID
3 mg BID

BID, twice daily; CA, cholate.

BI 685509 selectively lowered the peripheral venous concentration of orally administered d4-Cholate, consistent with a selective effect of enhanced uptake of cholate by the liver from the portal circulation

Figure 3: Effect of 3 mg BID BI 685509 on serum concentrations of cholates

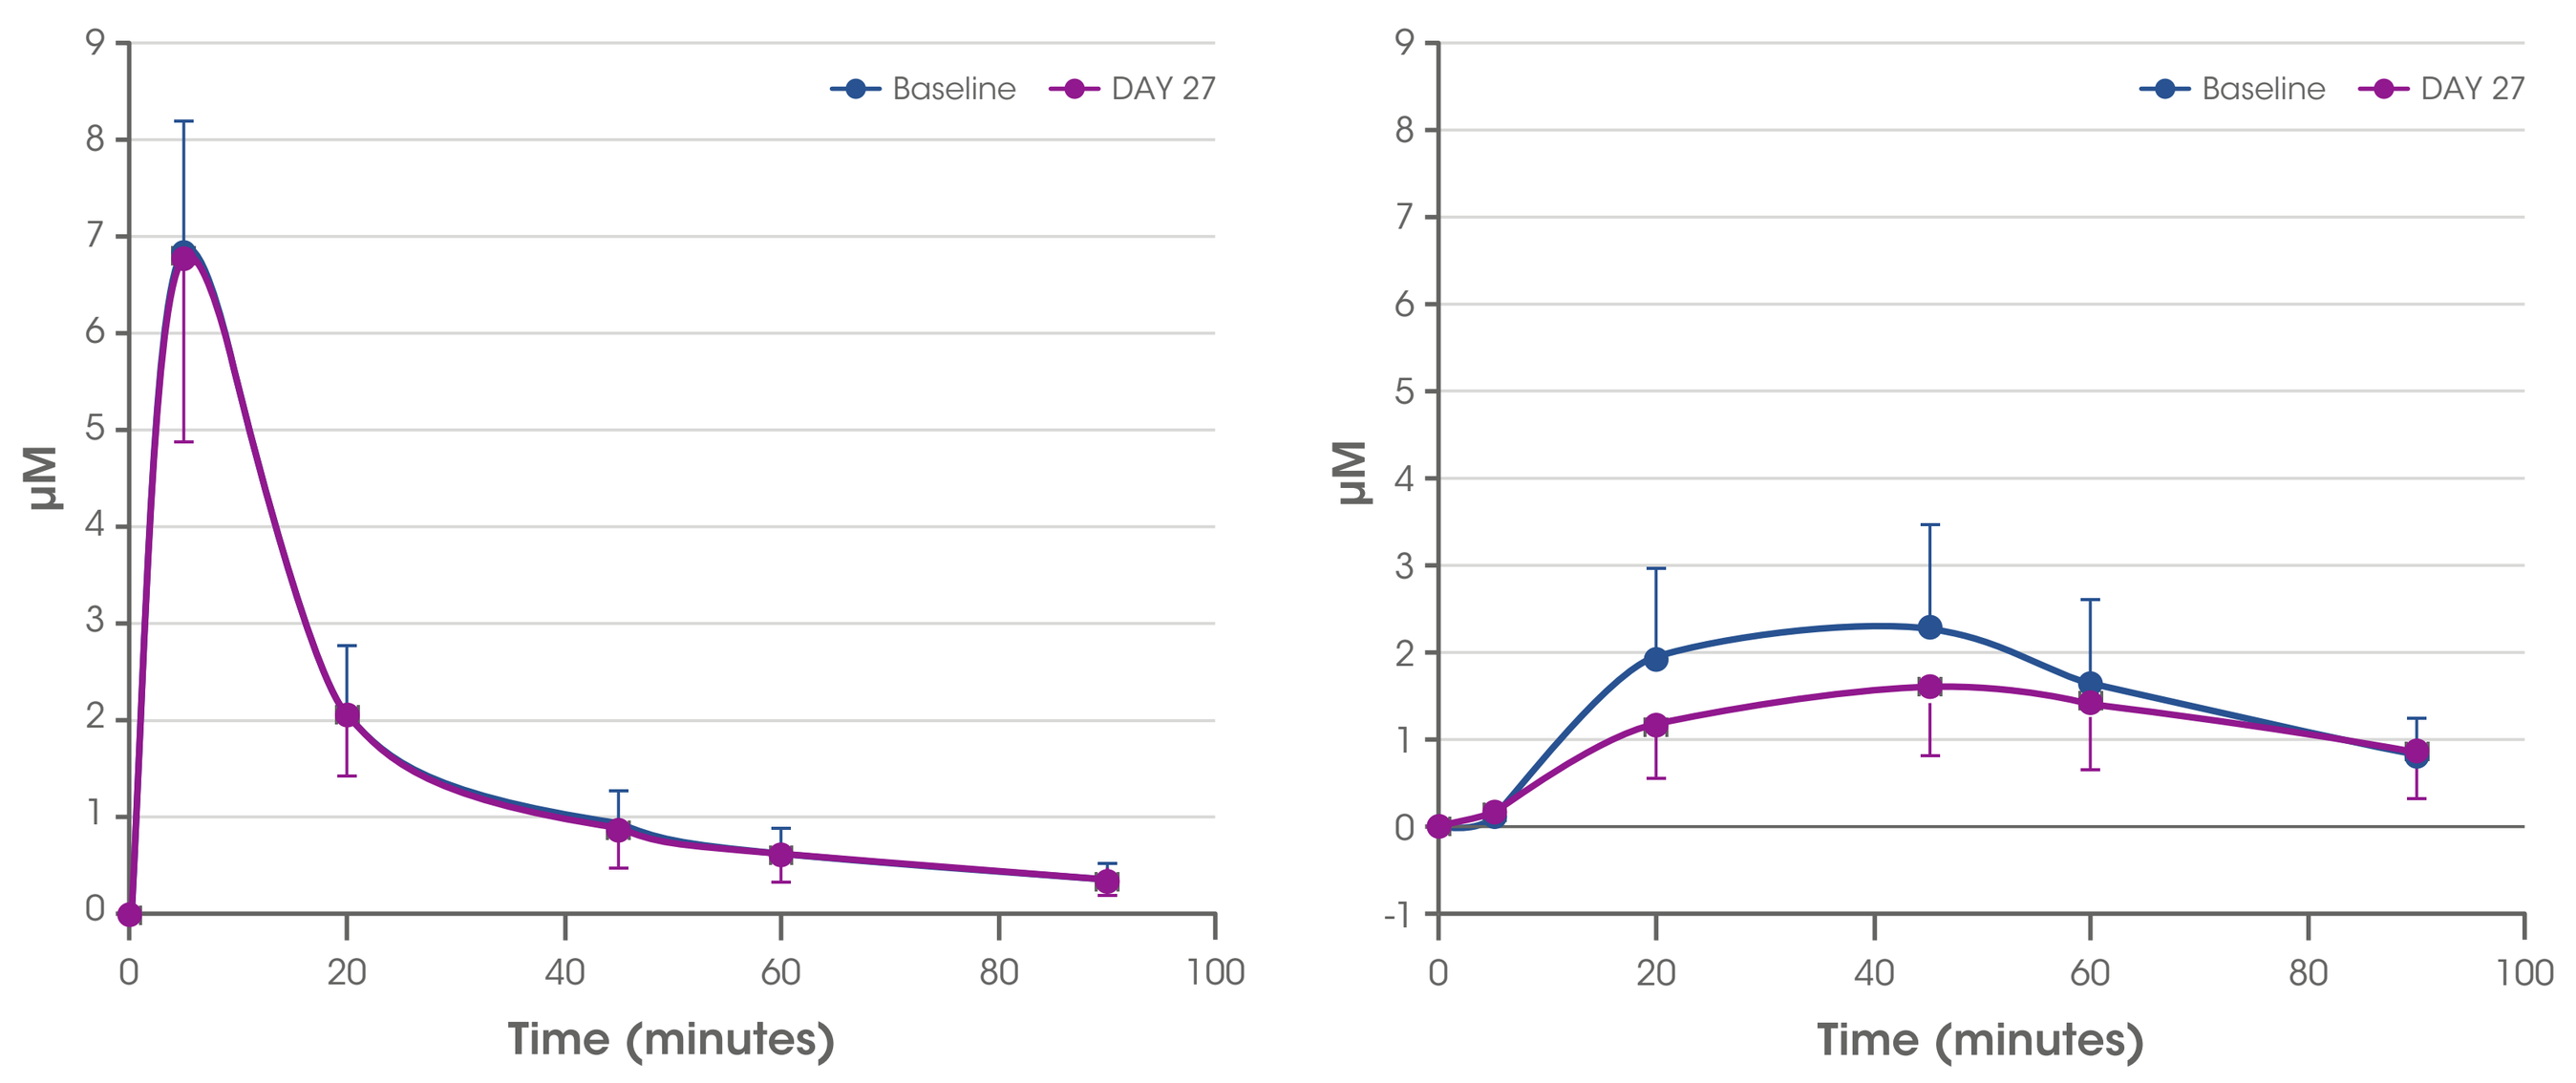
Intravenously administered [13C-Cholate]
Baseline
DAY 27
µM
Time (minutes)
0
20
40
60
80
100
Orally administered [d4-Cholate]
Baseline
DAY 27
µM
Time (minutes)

		Systemic HFR	Portal HFR	SHUNT %	Hepatic reserve	DSI
Baseline	Mean SD	2.96 0.54	6.81 3.21	49.0 14.8	45.9 10.7	27.1 5.4
DAY 27	Mean SD	3.10 0.91	8.22 4.02	42.0 12.3	49.4 11.9	25.3 6.0

BID, twice daily; DSI, disease severity index; HFR, hepatic filtration rate; SD, standard deviation.

Table 2. Baseline HepQuant results

- The subjects treated with BI 685509 had worse hepatic function (lower HFRs, lower HR, and higher DSI) and greater portal-systemic shunting (higher SHUNT) compared to subjects treated with placebo

Baseline HepQuant test parameter mean (SD)	1 mg BID (n=6)	2 mg BID (n=6)	3 mg BID (n=6)	Placebo (n=5)
SYST HFR, mL/min/kg	3.06 (0.64)	3.12 (0.75)	2.96 (0.54)	3.71 (0.98)
PORT HFR, mL/min/kg	7.23 (3.34)	7.44 (3.07)	6.81 (3.21)	10.87 (5.74)
SHUNT, %	47.9 (15.0)	46.9 (15.6)	49.0 (14.8)	38.6 (11.7)
DSI	26.4 (6.2)	26.0 (5.7)	27.1 (5.4)	21.7 (6.1)
STAT, µM	1.69 (1.07)	1.79 (0.87)	1.95 (0.88)	1.18 (0.88)
HR, %	47.1 (12.4)	48.0 (11.3)	45.9 (10.7)	56.5 (12.2)

BID, twice daily; DSI, disease severity index; HFR, hepatic filtration rate; HR, hepatic reserve; PORT, portal; QD, oce daily; SD, standard deviation; SYST, systemic.

Table 3. Change in HepQuant results from baseline to Day 27

- The reduction in SHUNT and increase in first-pass extraction of d4-Cholate (1-SHUNT) in the subjects on 3 mg BID BI 685509 was due to enhanced clearance of cholate from the portal circulation (increase in portal hepatic filtration rate, Port HFR)

Change in HepQuant test parameter mean (SD)	1 mg BID (n=6)	2 mg BID (n=6)	3 mg BID (n=6)	Placebo (n=5)
SYST HFR, mL/min/kg	0.12 (0.41)	-0.34 (0.70)	0.14 (0.53)	-0.18 (0.50)
PORT HFR, mL/min/kg	0.35 (2.67)	-0.01 (0.77)	**1.41 (1.03)**	-0.84 (4.32)
SHUNT, %	1.0 (7.0)	**-5.9 (6.1)**	**-7.1 (5.3)**	-1.2 (7.5)
DSI	-0.3 (3.1)	0.5 (2.1)	**-1.8 (1.8)**	0.5 (3.3)
STAT, µM	-0.02 (0.33)	**-0.26 (0.54)**	**-0.21 (0.84)**	0.01 (0.46)
HR, %	0.6 (6.2)	-1.0 (4.3)	**3.5 (3.7)**	-1.0 (6.6)

BID, twice daily; DSI, disease severity index; HFR, hepatic filtration rate; HR, hepatic reserve; PORT, portal; QD, oce daily; SD, standard deviation; SYST, systemic.
Values highlighted in the table represent significant/near significant changes: PORT HFR and SHUNT: $p<0.05$, DSI and HR: $0.05<p<0.10$

Conclusions

- The results from this subgroup analysis indicate that BI 685509 improved hepatic perfusion in subjects with CP-A cirrhosis. In addition, first-pass extraction of d4-CA was increased with BI 685509 in a dose-dependent manner, with a significant increase observed with 3 mg BID (p=0.02)
- The HepQuant SHUNT test successfully defined differences in baseline disease severity between dose groups, detecting early treatment effects and defining dose response using small sample sizes

Disclosures

This work was funded by Boehringer Ingelheim. JE, CS, and IG are employees of Boehringer Ingelheim. SMH and GTE are employees of HepQuant, LLC. EL reports research/grant support from 89Bio, Allergan Inc, Akero Therapeutics, AstraZeneca, Axcella Health, Bristol Myers Squibb, Boehringer Ingelheim, Celgene, Durect Corporation, Eli Lilly and Company, Elobix, Enanta Pharmaceuticals, Enyo, Galmed Pharmaceuticals, Genfit, Gilead Sciences, Hanmi Pharmaceuticals, Intercept Pharmaceuticals, Laboratory for Advanced Medicine, Madrigal Pharmaceuticals, Merck, Metacrine Novartis, Novo Nordisk, Octeta Therapeutics, Poxel, Roche Pharmaceuticals, Viking, and Zydus. Medical writing support was provided by Susie Eaton, MBio, of OPEN Health Communications (London, UK) and was funded by Boehringer Ingelheim.

Poster: American Association for the Study of Liver Diseases (AASLD) The Liver Meeting®, November 12–15, Anaheim, CA, USA
Presenting author: Eric Lawitz, Texas Liver Institute, University of Texas, San Antonio, USA; Email: lawitz@txliver.com

References:

1. Derbyshire ER, Marletta MA. *Annu Rev Biochem* 2012;81:533-559.
2. Budworth J, et al. *Biochem Biophys Res Commun* 1999;263:696-701.
3. Zhao Y, et al. *Proc Natl Acad Sci U S A* 1999;96:14753-14758.
4. Wiest R, Groszmann RJ. *Hepatology* 2002;35:478-491.
5. Sarela AI, et al. *Gut* 1999;44:749-753.
6. Morales-Ruiz M, et al. In: Lanzer P, editor. PanVascular Medicine. Berlin, Heidelberg: Springer Berlin Heidelberg; 2015. p. 3631-3665.
7. Langer DA, Shah VH. *J Hepatol* 2006;44:209-216.
8. Burton JR, Jr.et al. *Transl Res* 2021;233:5-15.
9. Wieland A, et al. *Clin Gastroenterol Hepatol* 2021; Epub ahead of print.